ERAS-601, a potent inhibitor of SHP2, synergistically enhances the activity of gilteritinib, a FLT3 inhibitor, in FLT3-mutated AML tumor models

Leenus Martin, Roopal Patel, Jingchuan Zhang, Robin Nevarez, Taylor Congdon, Gary Chiang, Les Brail, Robert Shoemaker. Erasca, San Diego, CA

ERASCA™

3345

Abstract

ERAS-601 is a potent, small molecule allosteric inhibitor of wildtype SHP2, a non-receptor protein tyrosine phosphatase (PTP) encoded by the *PTPN11* gene. ERAS-601 inhibits wildtype SHP2 biochemically with an IC50 of 4.6 nM and demonstrates selectivity across panels of 300 kinases and 12 phosphatases. SHP2 mediates upstream receptor tyrosine kinase (RTK) signaling via its phosphatase-mediated regulation of guanine nucleotide exchange factors (GEFs). ERAS-601 inhibits the SHP2 dependent cycling of KRAS from the inactive GDP-bound state to the active GTP-bound state and demonstrates anti-proliferative activity in FLT3-mutated human cancer cells. Gilteritinib is a second-generation FLT3 inhibitor and has demonstrated clinical activity, but emerging resistance limits the benefit of monotherapy. Here, we explored the combination of gilteritinib with ERAS-601 in FLT3 mutated AML nonclinical models. The combination of ERAS-601 with gilteritinib inhibits the oncogenic RAS/MAPK signaling as measured by pERK1/2 and synergistically inhibits the cellular viability of FLT3-mutated AML cells in vitro. The combination of ERAS-601 with gilteritinib achieves a more durable tumor growth inhibition than the respective gilteritinib and ERAS-601 monotherapies in vivo. These nonclinical data support the clinical development of ERAS-601 in combination with gilteritinib in FLT3-altered AML in the HERKULES-4 Phase 1b/2 master protocol.

Introduction

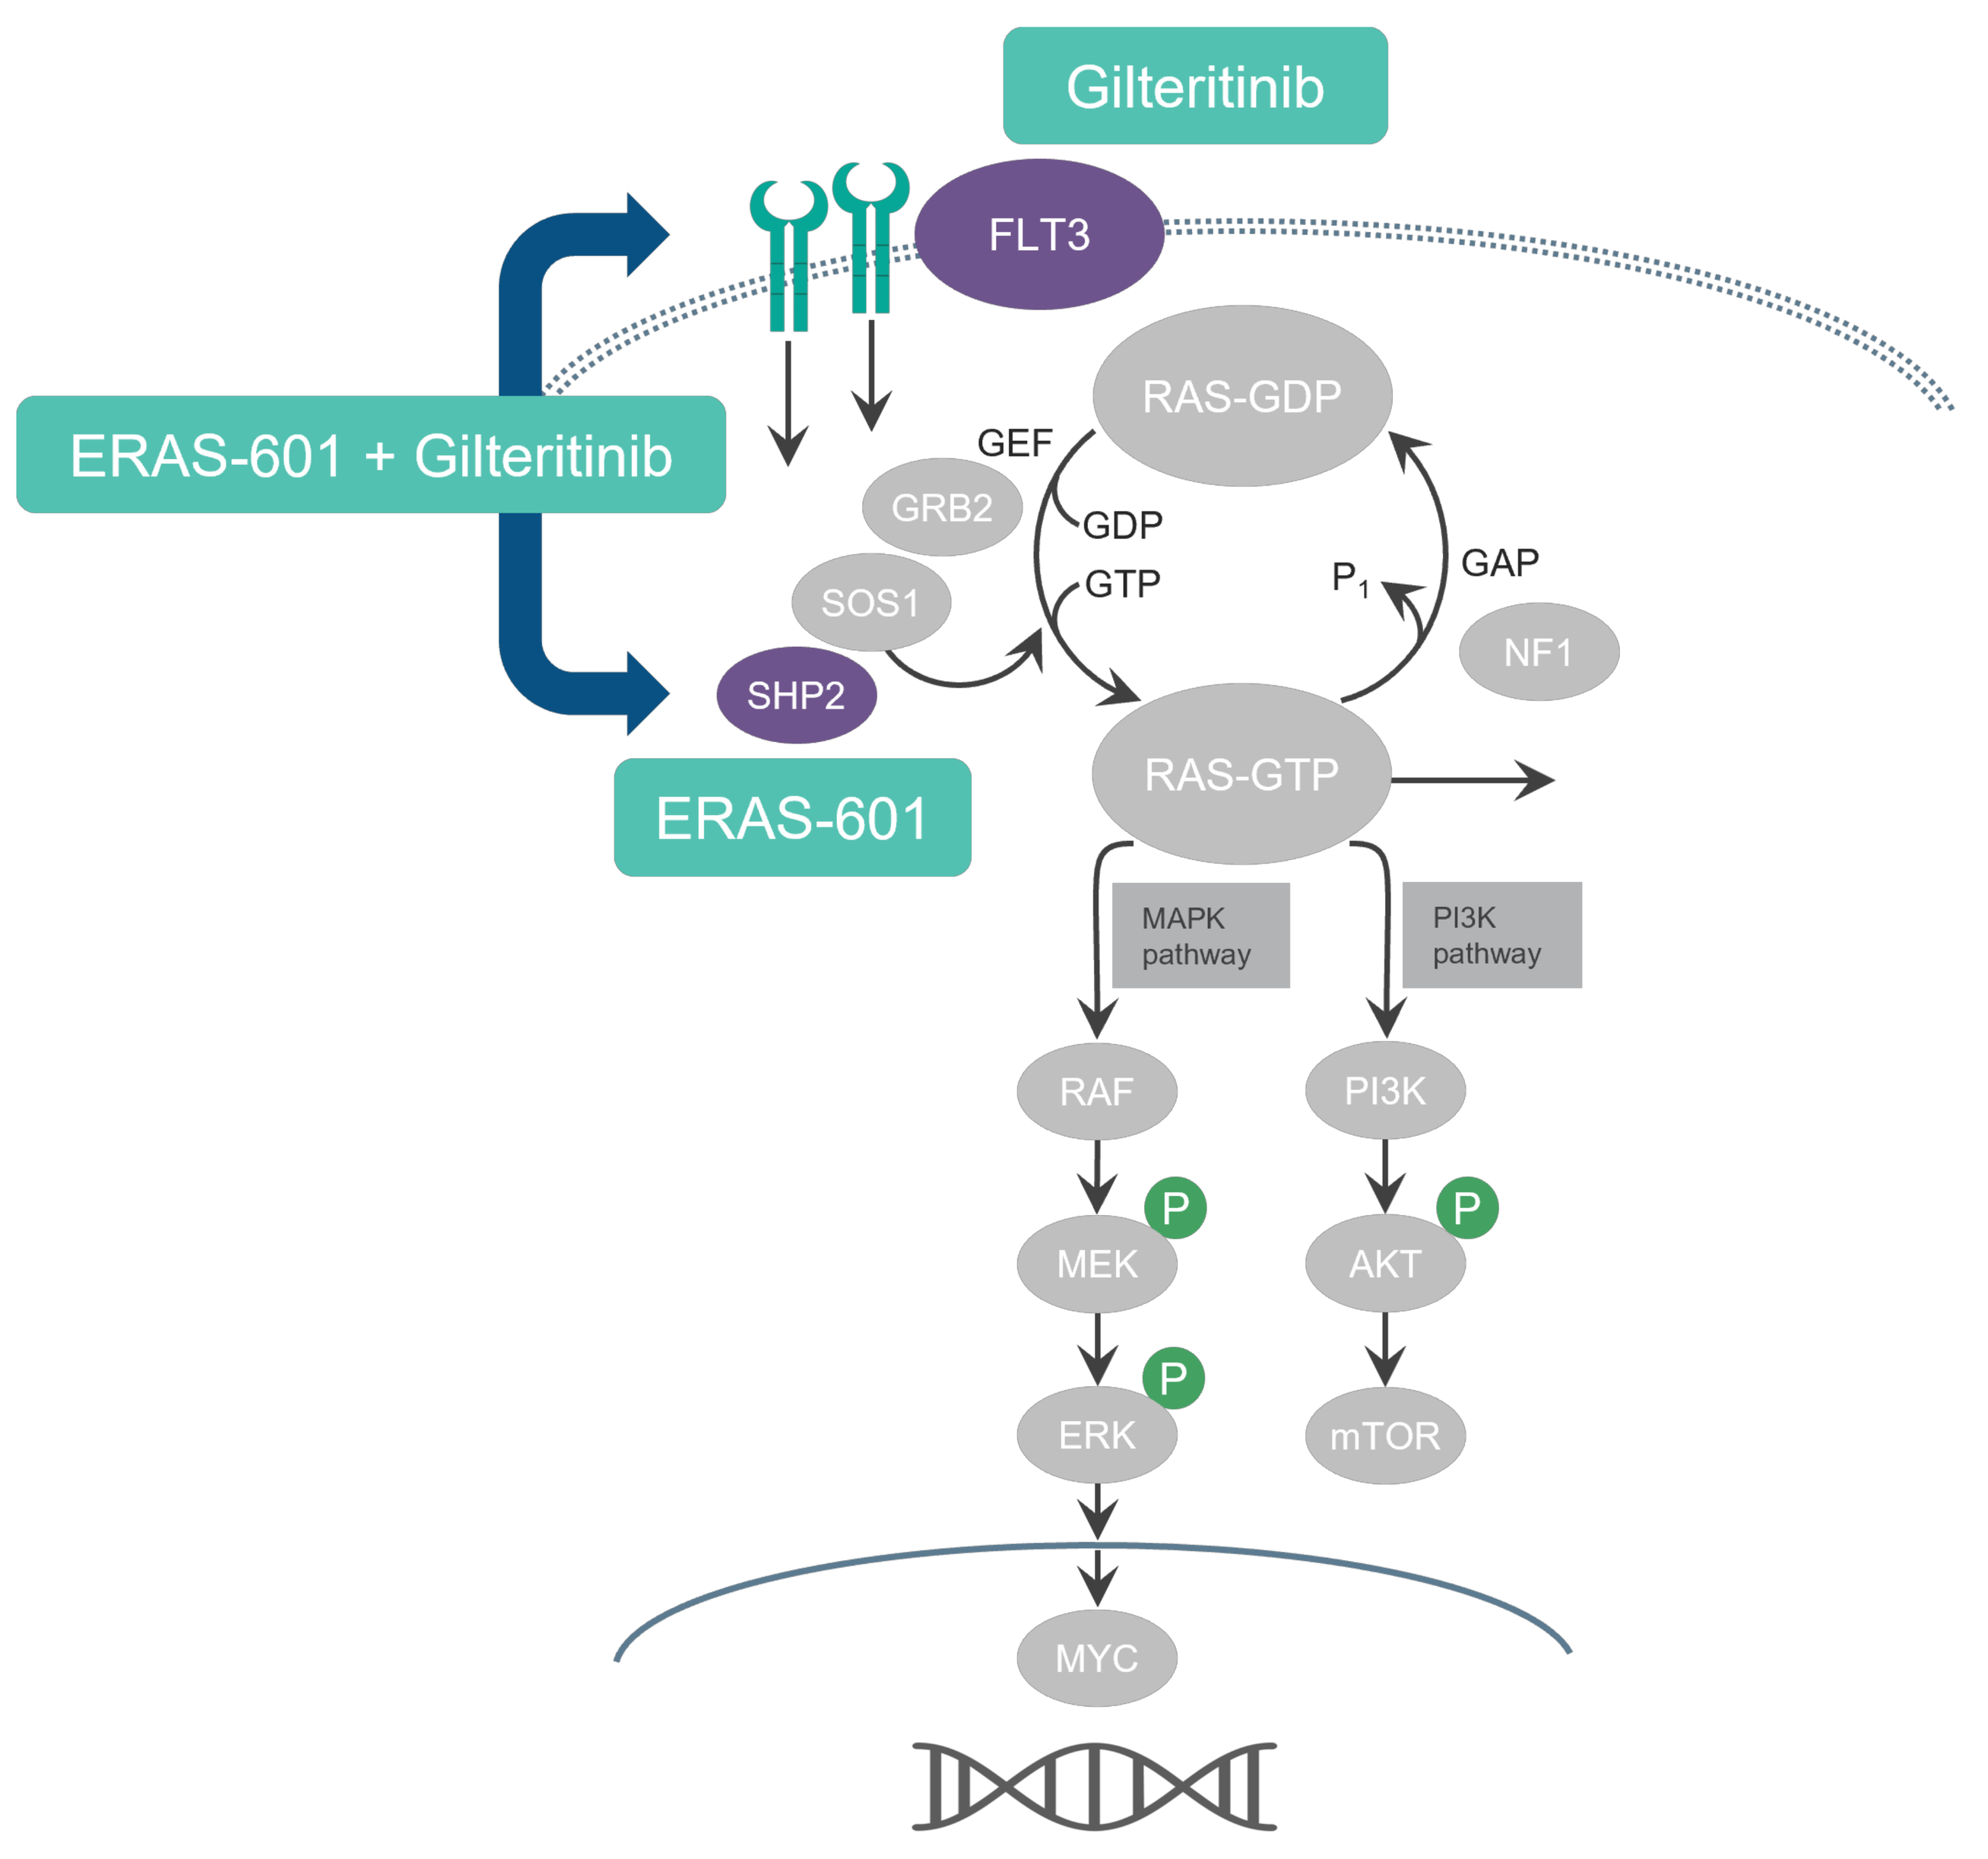
Gilteritinib
FLT3
ERAS-601 + Gilteritinib
RAS-GDP
GEF
GRB2
GDP
GTP
SOS1
P1
GAP
NF1
SHP2
ERAS-601
RAS-GTP
MAPK pathway
PI3K pathway
RAF
PI3K
P
MEK
AKT
ERK
mTOR
MYC

Figure 1. Schematic representation of ERAS-601 with gilteritinib combination

Results

ERAS-601 is a potent and selective small molecule allosteric inhibitor of SHP2

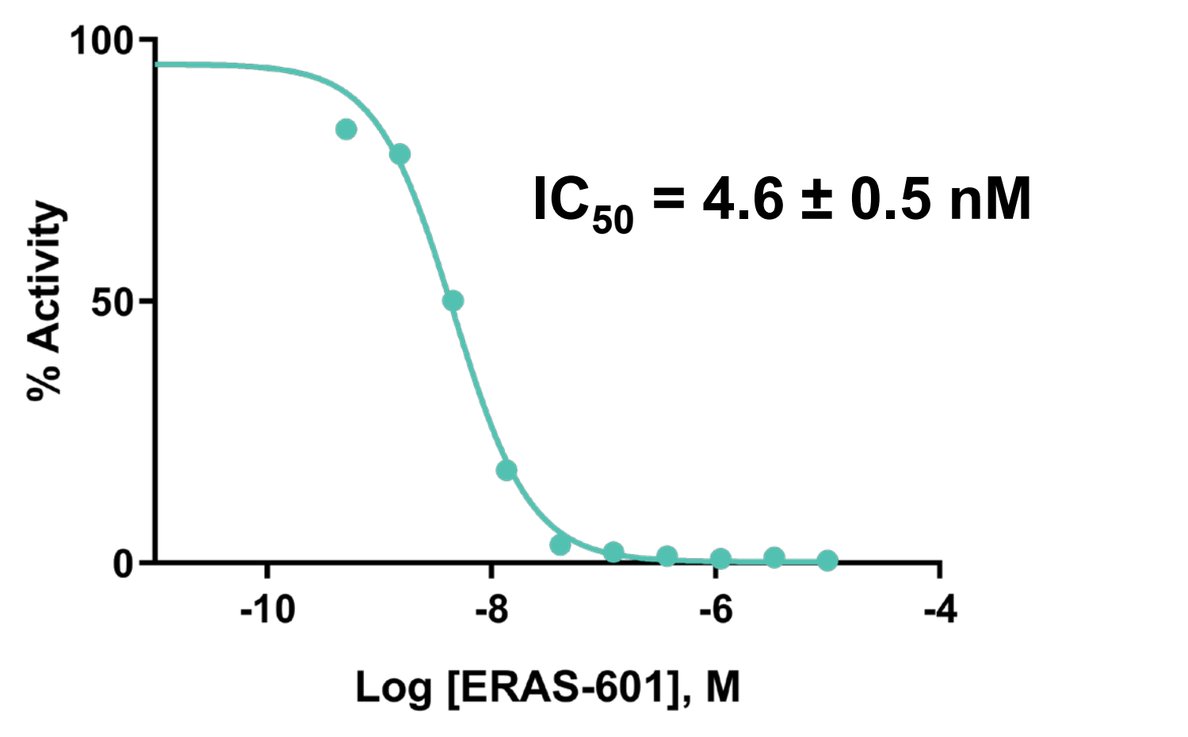
IC_{50} = 4.6 ± 0.5 nM
% Activity
Log [ERAS-601], M

PTPase	% Inhibition at 10 µM ERAS-601
PTPN11/SHP2 full length (on-target)	100%
PTPN11/SHP2 catalytic domain	0%
PTPRC/CD45	6%
PTPN2/TC-PTP	3%
PP2A Alpha/ PP2R1A	1%
PTPN6/SHP1	0%
PTPN1/PTP1B-CD	0%
PTPN12/PTP-PEST	0%
PP1B	0%
DUSP22/MKPX	0%
PTPN7/LC-PTP	0%
PP1A	0%

Figure 2. Biochemical activity of ERAS-601. In vitro phosphatase activity of full-length SHP2 was assessed in the presence of increasing concentrations of ERAS-601. The IC50 value shown is the average of 2 replicates. For selectivity, ERAS-601 was assessed in vitro against a panel of protein tyrosine phosphatases at 10 µM and no off-target activity at >6% at 10 µM was observed. ERAS-601 also showed no appreciable inhibition against any off-target kinase in a 300 kinase panel (<30% inhibition @ 1 µM).

The combination of ERAS-601 with gilteritinib synergistically inhibited cellular viability in FLT3-ITD mutated AML cells

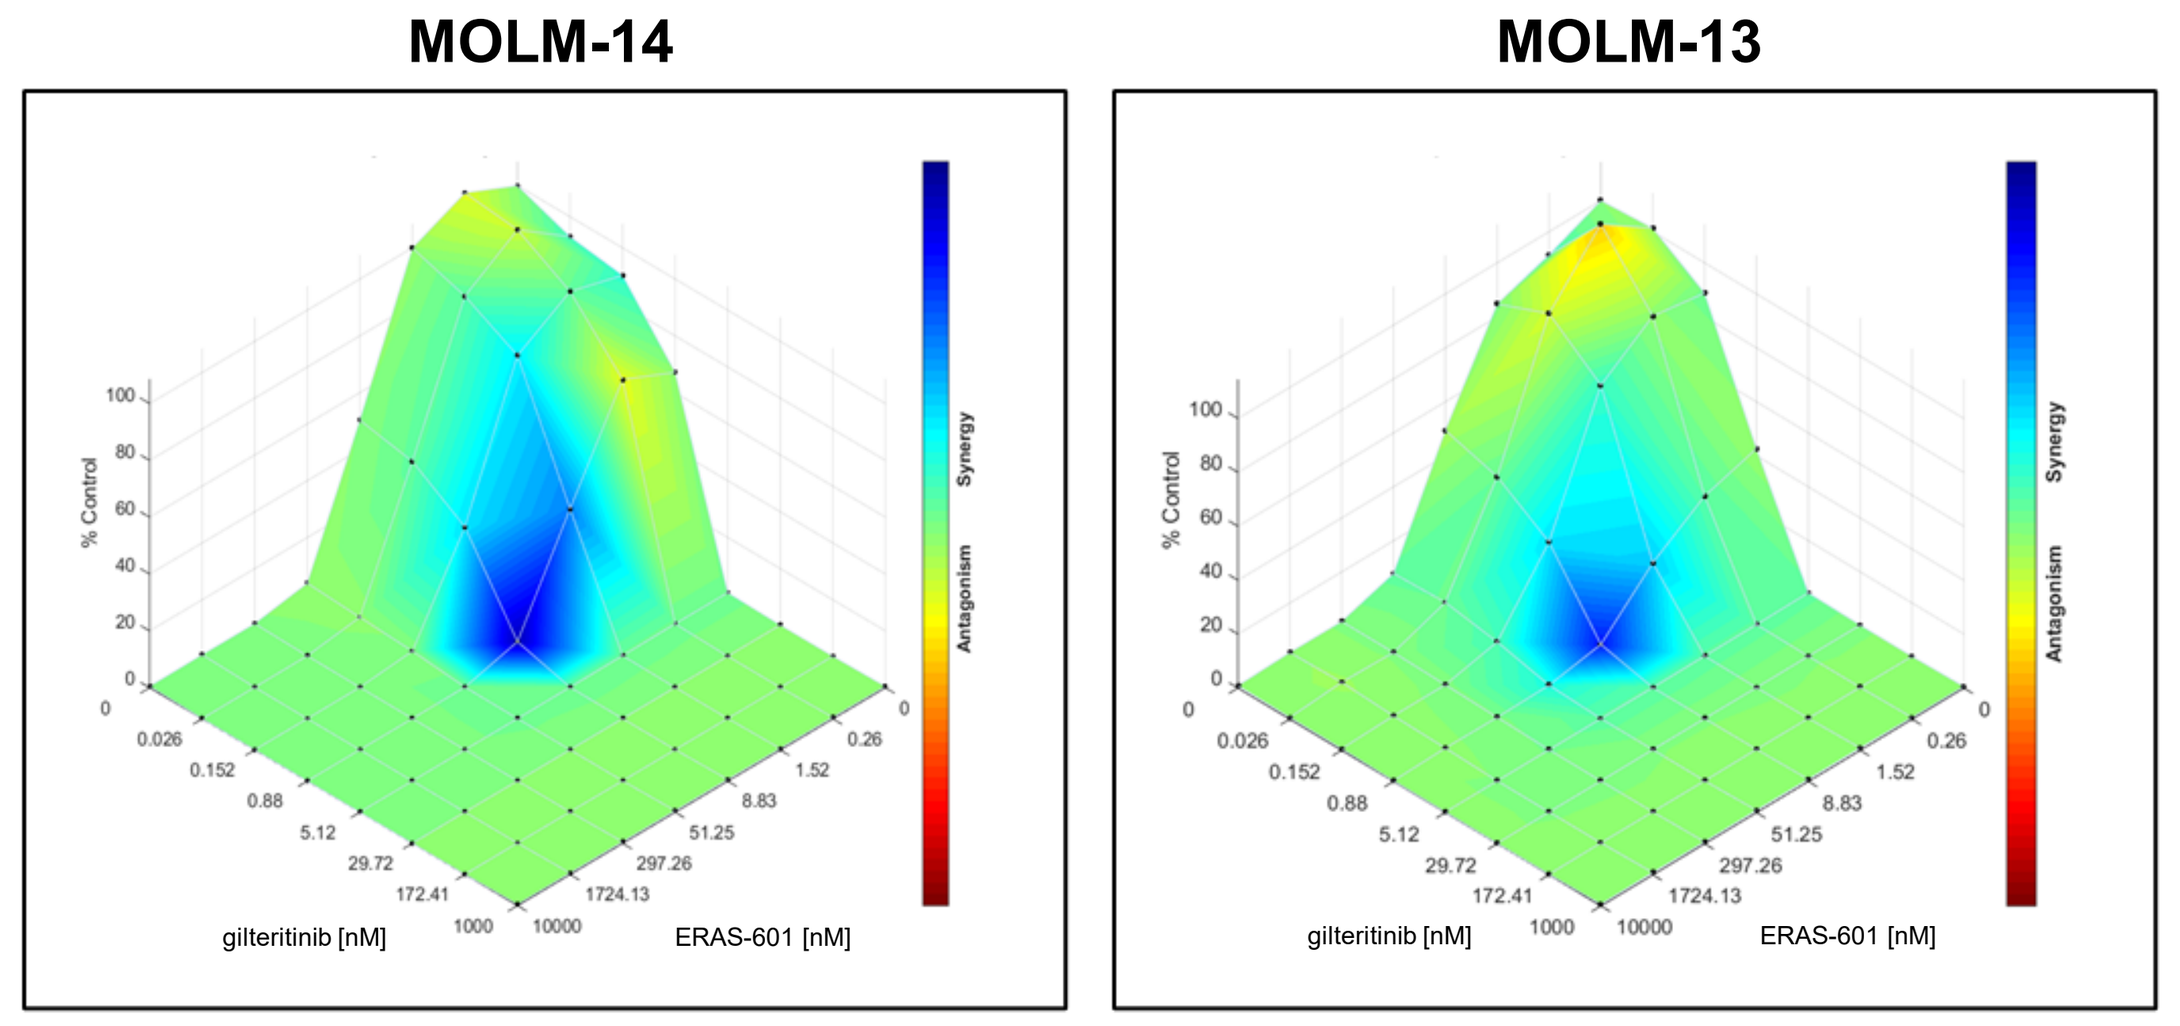
MOLM-14
MOLM-13
% Control
gilteritinib [nM]
ERAS-601 [nM]
Synergy
Antagonism

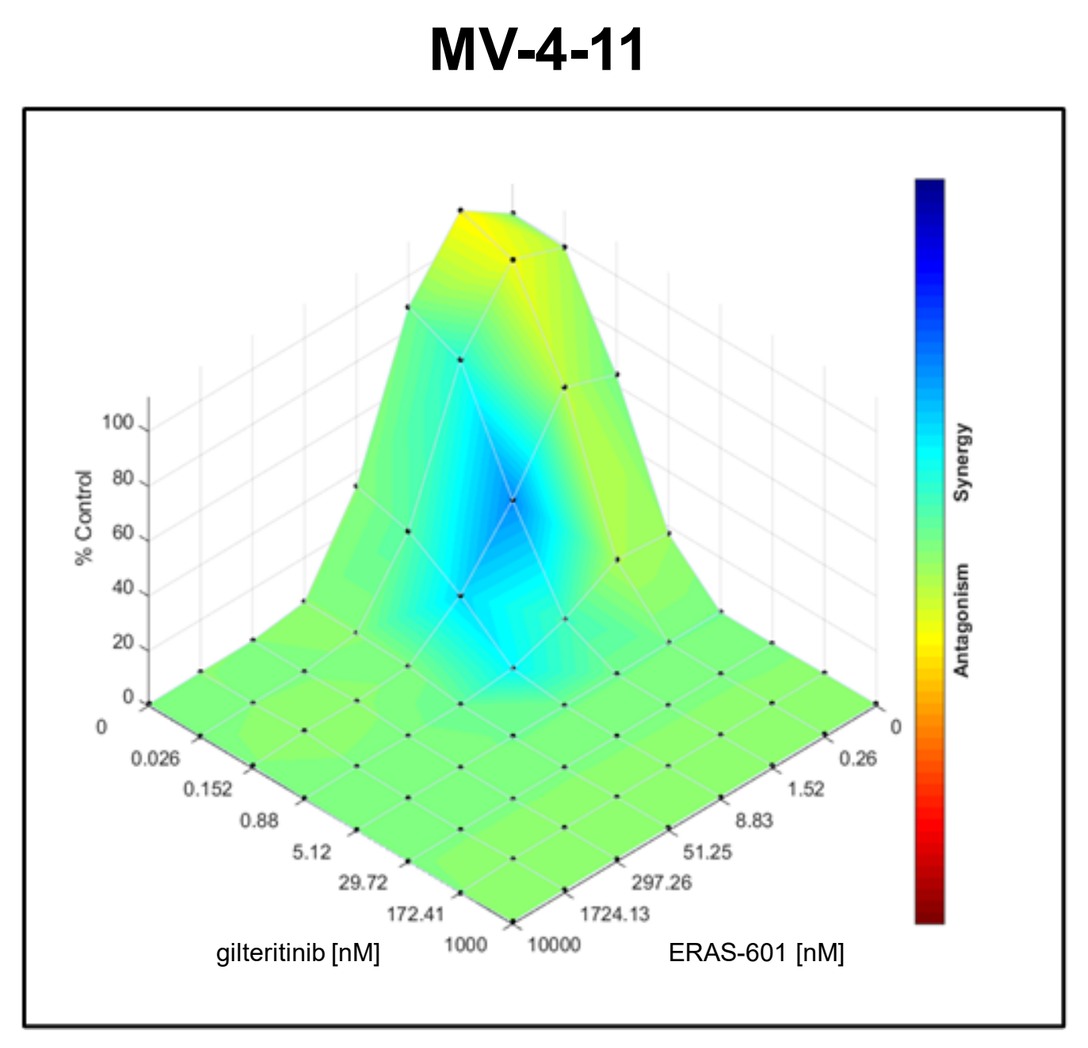
MV-4-11
% Control
gilteritinib [nM]
ERAS-601 [nM]
Synergy
Antagonism

Figure 3. 2D surface plots of the ERAS-601 with gilteritinib combination in FLT3-ITD cell lines. MOLM-14, MOLM-13 and MV-4-11 cell lines were treated with ERAS-601 and gilteritinib. Cell proliferation was assessed in a 5 day 2D CellTiter-Glo assay and HSA synergy plots were generated using Combenefit software.

The combination of ERAS-601 with gilteritinib showed enhanced inhibition of ERK1/2 phosphorylation in FLT3-ITD mutated MOLM-14 cells

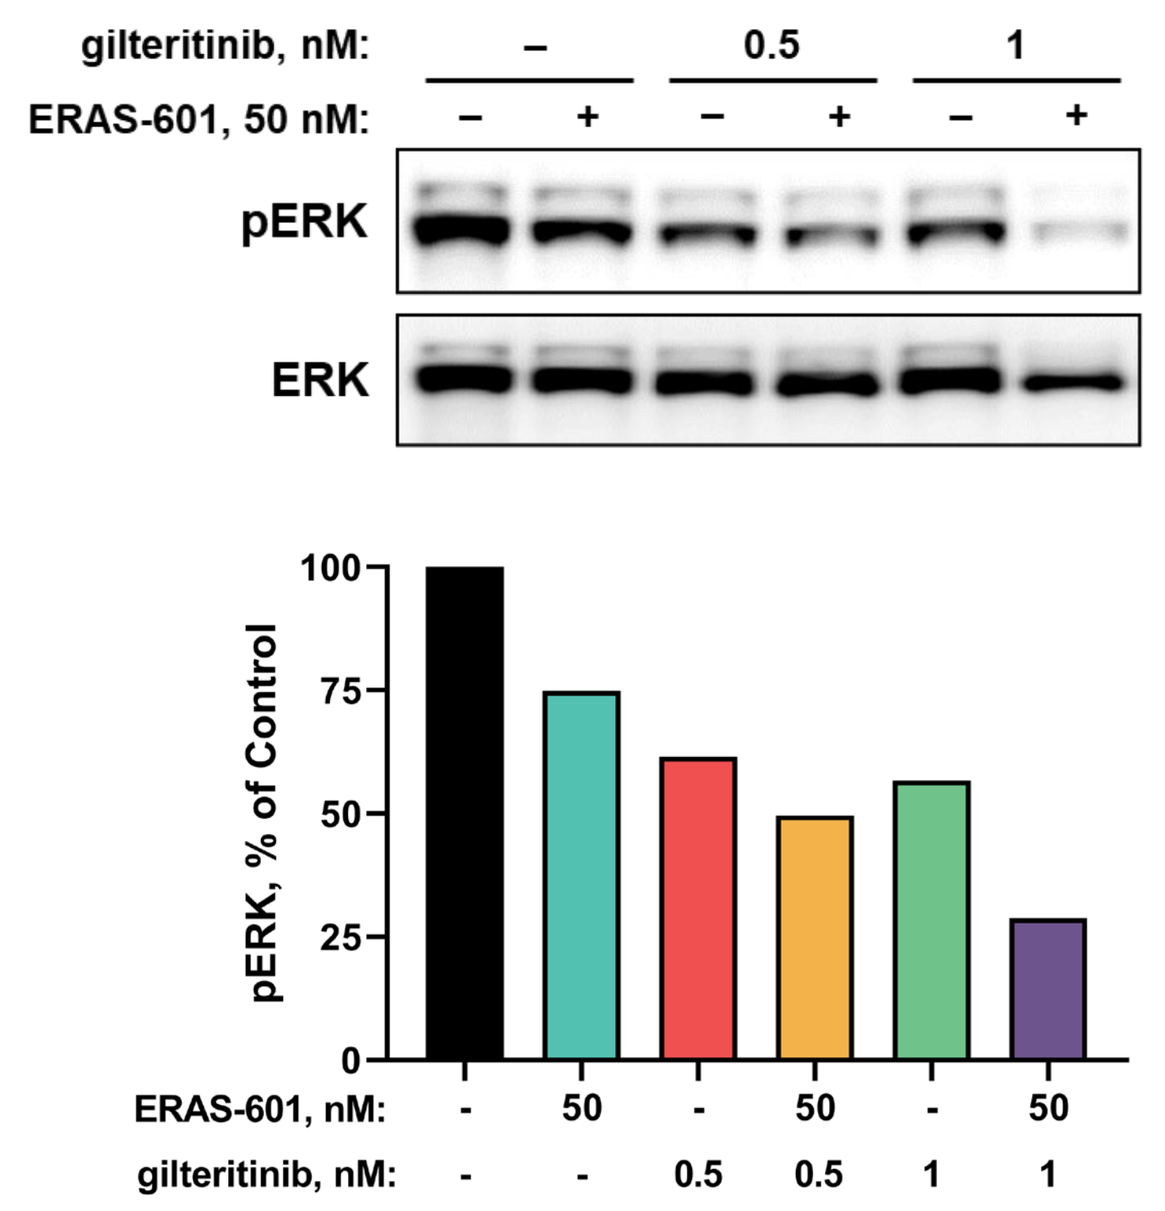
gilteritinib, nM:
ERAS-601, 50 nM:
pERK
ERK
pERK, % of Control
ERAS-601, nM:
gilteritinib, nM:

Figure 4. Assessment of ERK1/2 signaling following ERAS-601 and gilteritinib treatment. The FLT3-ITD mutated AML cell line MOLM-14 was treated with ERAS-601 alone and ERAS-601 with gilteritinib combination for 6 hours and cell lysates were harvested. *Top panel*, immunoblot of pERK1/2 and total ERK. *Bottom panel*, quantitation plot of pERK1/2 after being normalized to total ERK.

The combination of ERAS-601 with gilteritinib showed enhanced inhibition of ERK1/2 phosphorylation in FLT3-ITD mutated MV-4-11 cells

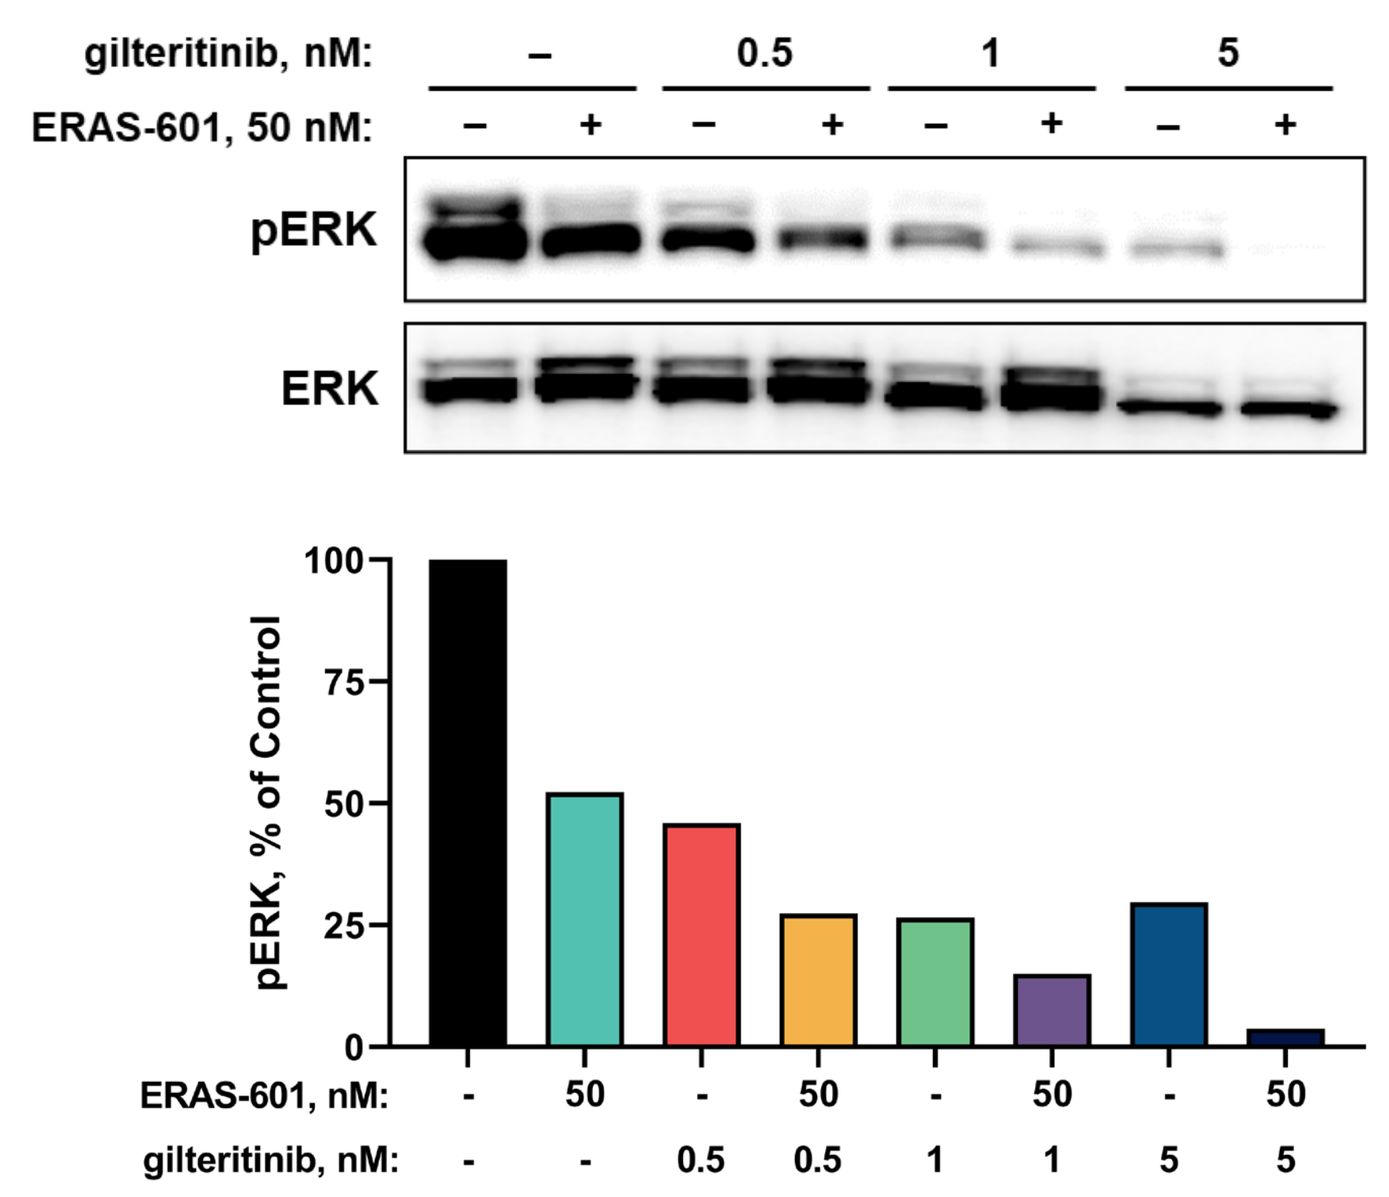
gilteritinib, nM:
ERAS-601, 50 nM:
pERK
ERK
pERK, % of Control
ERAS-601, nM:
gilteritinib, nM:

Figure 5. Assessment of ERK1/2 signaling following ERAS-601 and gilteritinib treatment. The FLT3-ITD mutated AML cell line MV-4-11 was treated with ERAS-601 alone and ERAS-601 with gilteritinib combination for 6 hours and cell lysates were harvested. *Top panel*, immunoblot of pERK1/2 and total ERK. *Bottom panel*, quantitation plot of pERK1/2 after being normalized to total ERK.

The combination of ERAS-601 with gilteritinib achieved durable tumor growth inhibition in a FLT3-ITD mutated AML subcutaneous CDX model MOLM-13

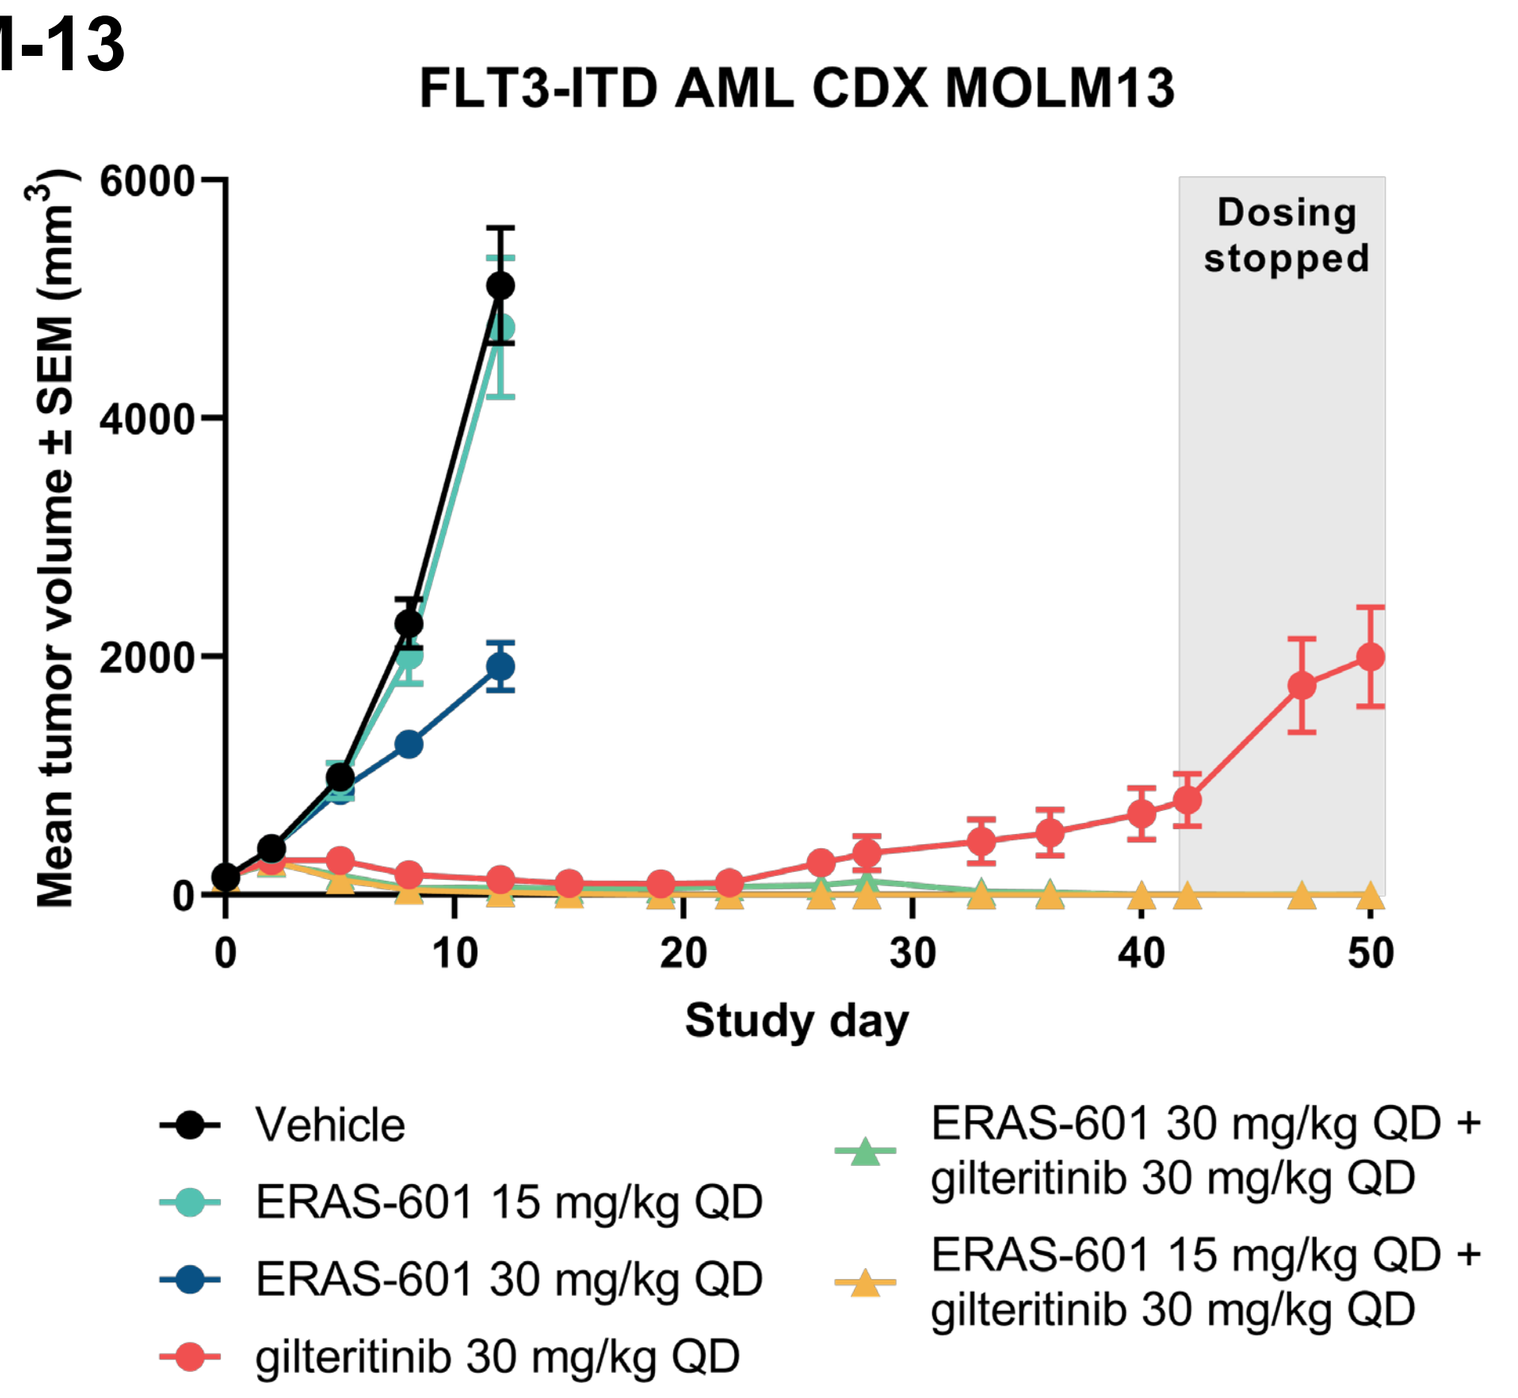
FLT3-ITD AML CDX MOLM13
Dosing stopped
Mean tumor volume ± SEM (mm³)
Study day
Vehicle
ERAS-601 15 mg/kg QD
ERAS-601 30 mg/kg QD
gilteritinib 30 mg/kg QD
ERAS-601 30 mg/kg QD + gilteritinib 30 mg/kg QD
ERAS-601 15 mg/kg QD + gilteritinib 30 mg/kg QD

Figure 6. In vivo efficacy of ERAS-601 with gilteritinib combination in a FLT3-ITD mutant subcutaneous xenograft. Immunodeficient mice bearing MOLM-13 tumor xenografts were orally dosed with indicated single agent and combination treatments. Tumors were measured on the indicated days and mean tumor volumes were plotted. *SEM*, standard error of the mean.

The combination of ERAS-601 with gilteritinib achieved superior tumor growth inhibition to respective ERAS-601 and gilteritinib monotherapies in a FLT3-ITD mutated AML engrafted CDX model MOLM-13-Luc

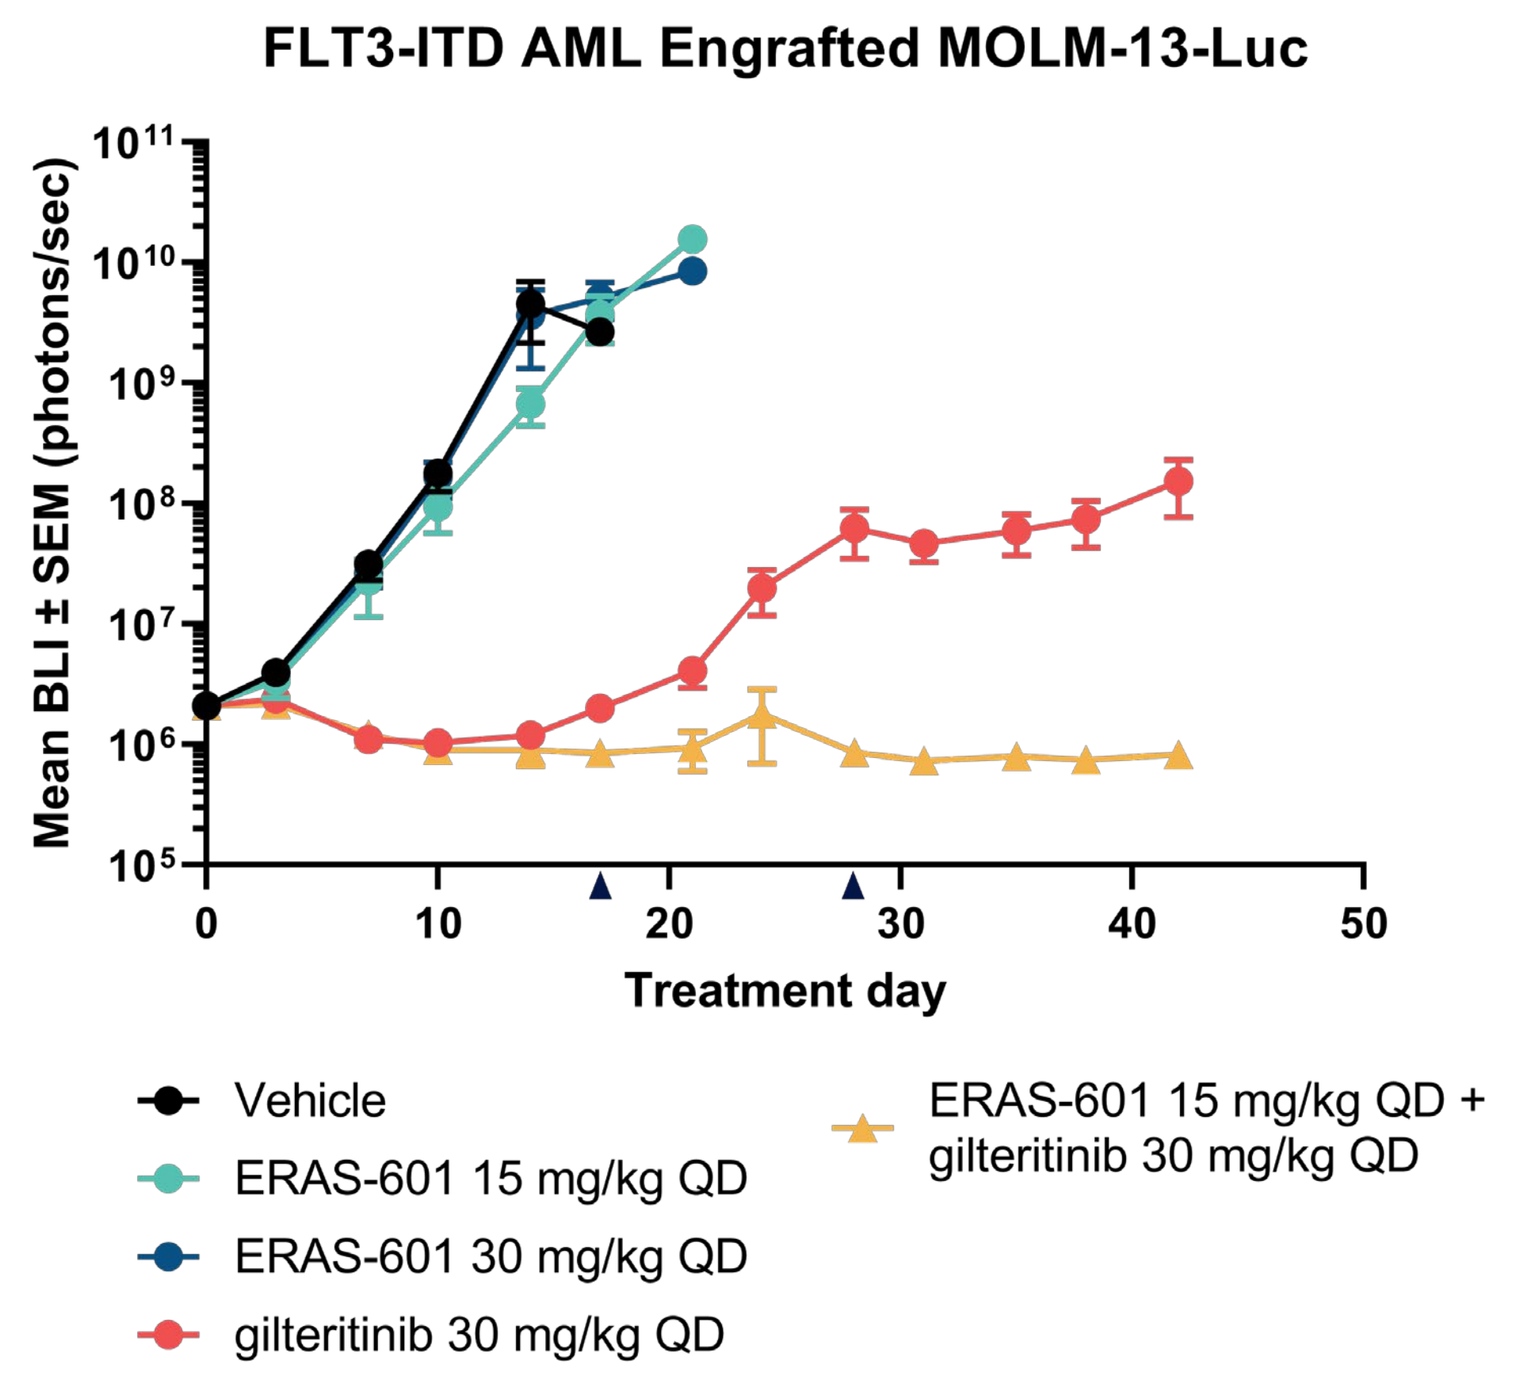
FLT3-ITD AML Engrafted MOLM-13-Luc
Mean BLI ± SEM (photons/sec)
Treatment day
Vehicle
ERAS-601 15 mg/kg QD
ERAS-601 30 mg/kg QD
gilteritinib 30 mg/kg QD
ERAS-601 15 mg/kg QD + gilteritinib 30 mg/kg QD

Figure 7. In vivo efficacy of ERAS-601 with gilteritinib combination in a FLT3-ITD mutant engraftment model. Immunodeficient mice harboring engrafted luciferase labeled MOLM-13 cells ("MOLM-13-Luc") were orally dosed with indicated single agent and combination treatments. Bioluminescence intensity (BLI) was measured following luciferin injection on the indicated days and mean BLI was plotted. Arrowheads denote imaging days; *SEM*, standard error of the mean.

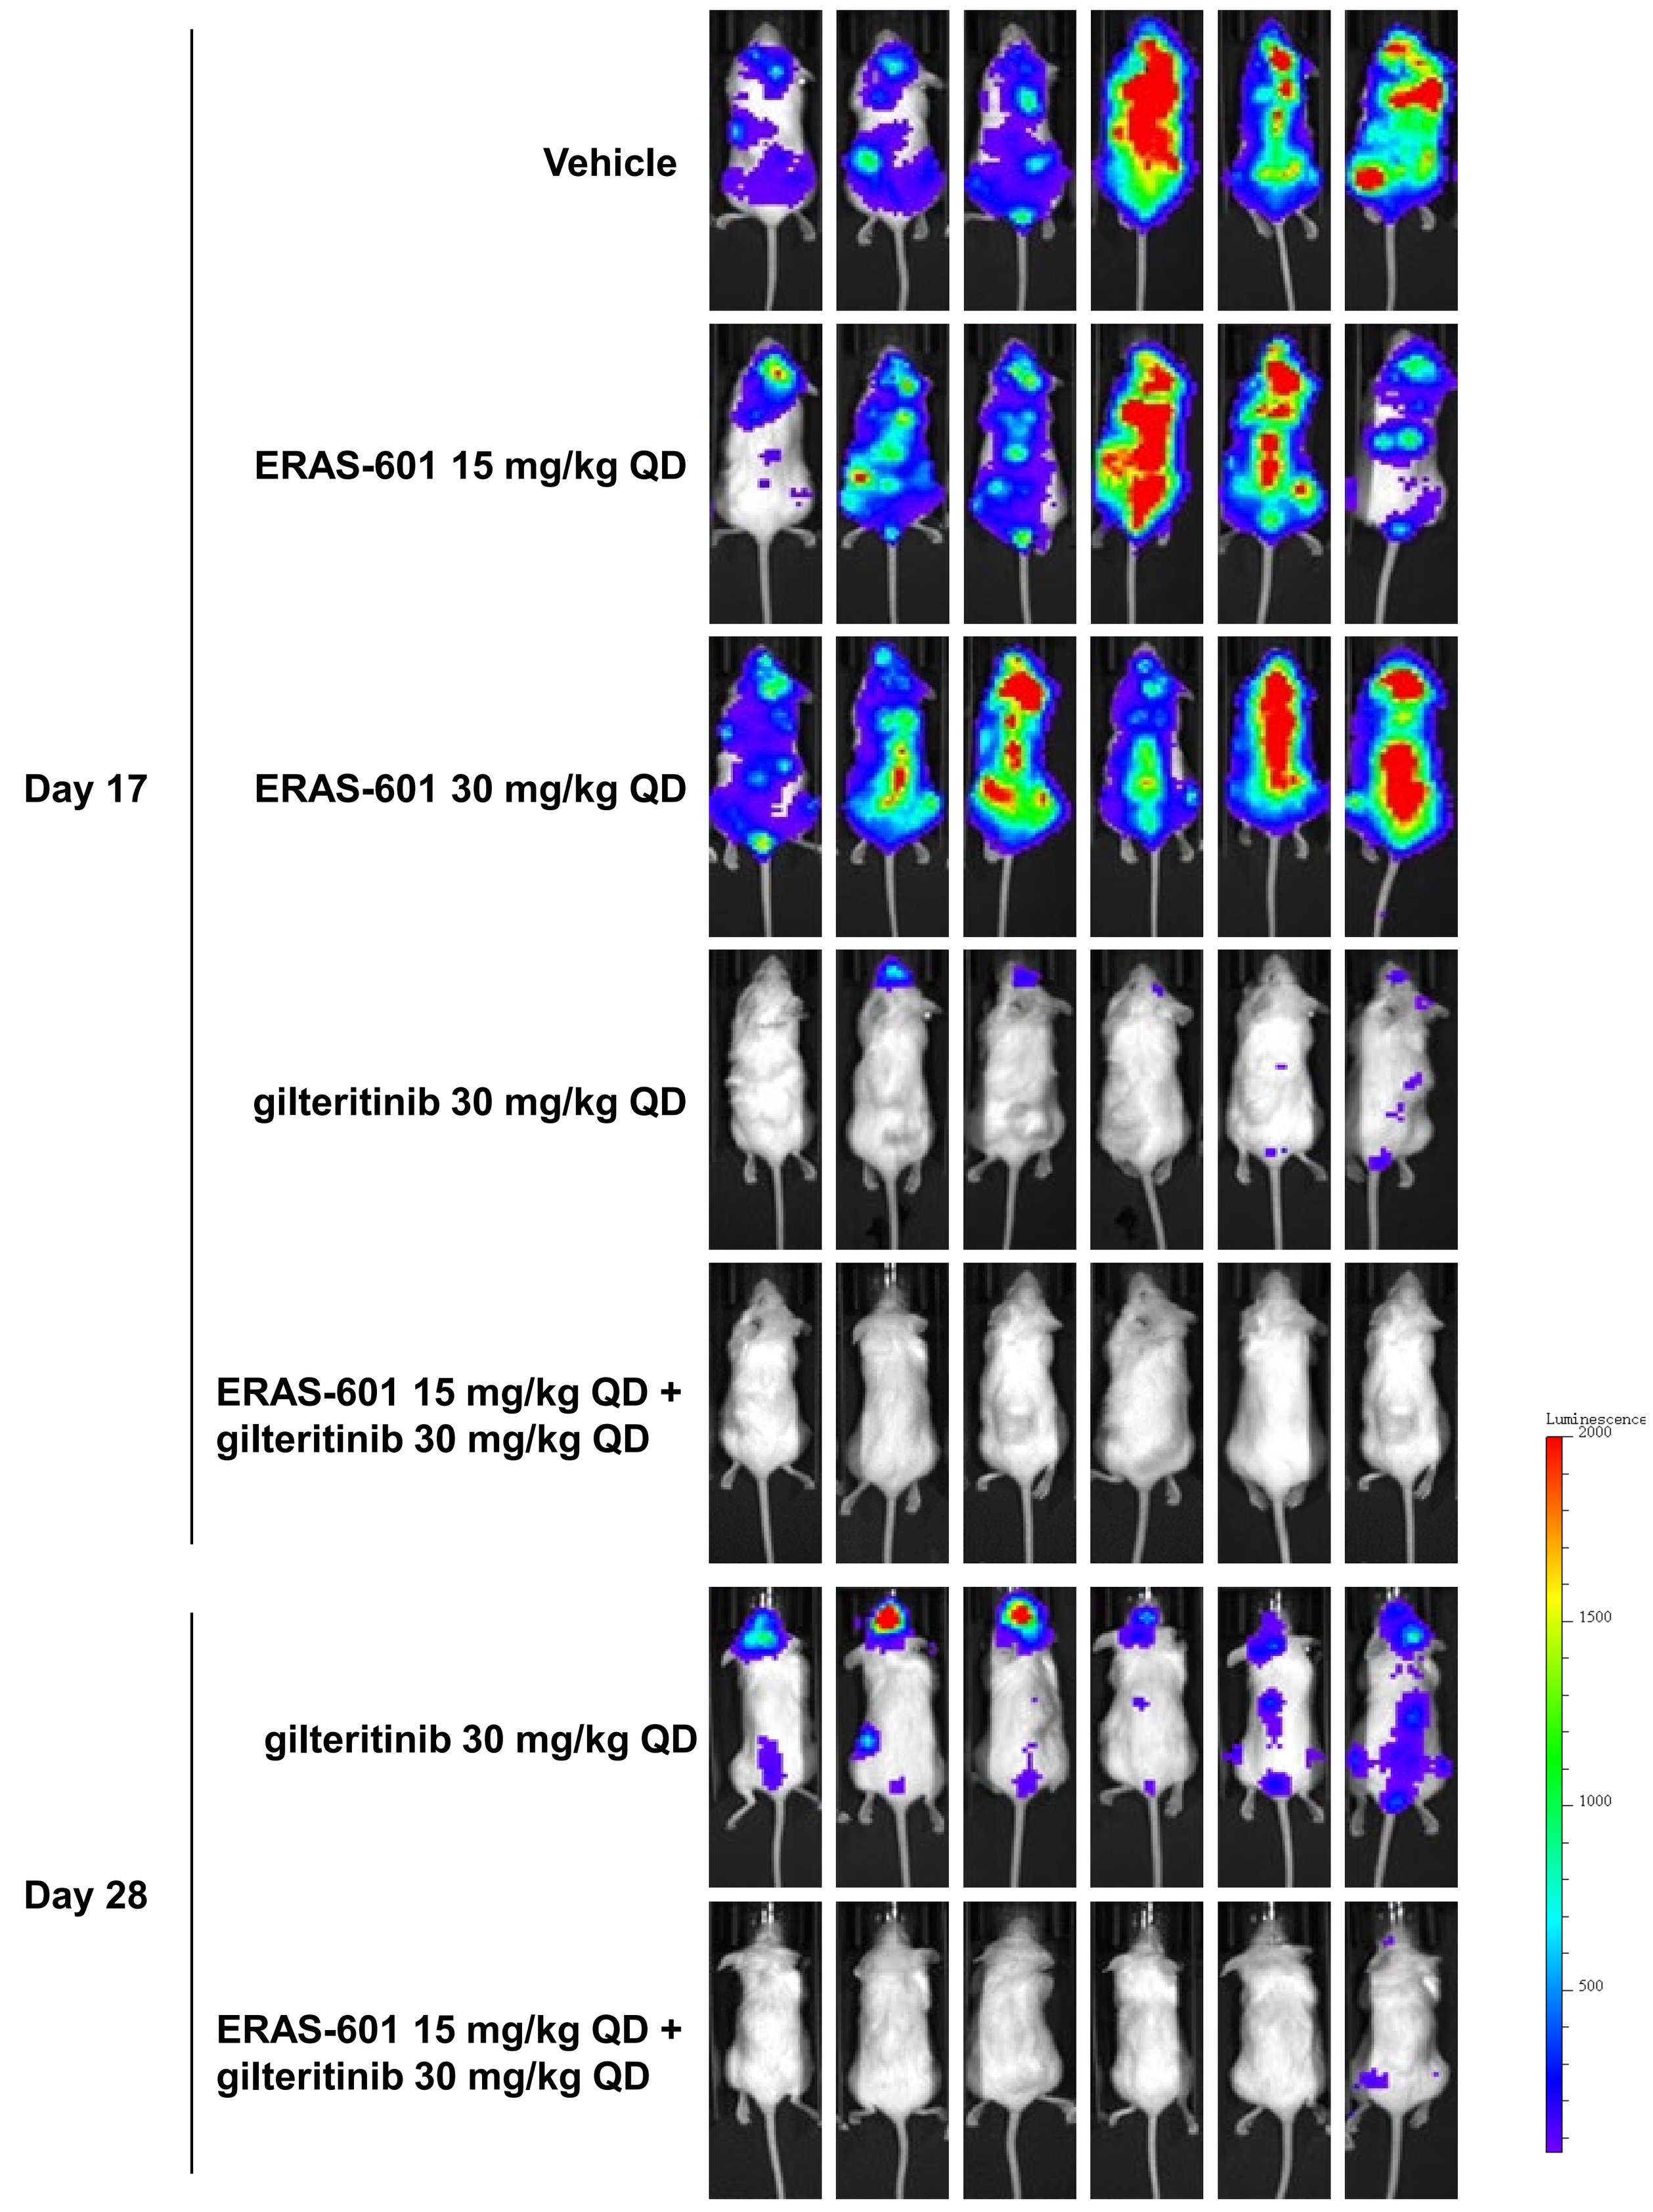
Day 17
Vehicle
ERAS-601 15 mg/kg QD
ERAS-601 30 mg/kg QD
gilteritinib 30 mg/kg QD
ERAS-601 15 mg/kg QD + gilteritinib 30 mg/kg QD
Day 28
gilteritinib 30 mg/kg QD
ERAS-601 15 mg/kg QD + gilteritinib 30 mg/kg QD
Luminescence

Figure 8. Bioluminescent images of ERAS-601 with gilteritinib combination efficacy in a FLT3-ITD mutant engraftment model. Immunodeficient mice harboring engrafted luciferase labeled MOLM-13 cells ("MOLM-13-Luc") were orally dosed with indicated single agent and combination treatments. The figure shows representative bioluminescence images of tumor-bearing mice at day 17 and day 28.

Conclusions

- The combination of ERAS-601 with gilteritinib inhibits RAS/MAPK pathway signaling in FLT3-ITD mutant cellular models in vitro as measured by pERK1/2 and cellular viability
- ERAS-601 with gilteritinib achieves more durable tumor growth inhibition than respective gilteritinib and ERAS-601 monotherapies in the FLT3-ITD mutant CDX AML MOLM-13 subcutaneous model
- The combination of ERAS-601 with gilteritinib achieves tumor growth inhibition that is superior to respective ERAS-601 and gilteritinib monotherapies in FLT3-ITD mutant MOLM13-Luc AML engraftment model
- These data support the clinical development of ERAS-601 in combination with gilteritinib in FLT3-altered AML in the HERKULES-4 Phase 1b/2 master protocol